Phase 1b/2a Study of PLX51107, a Small Molecule BET Inhibitor, in Subjects with Advanced Hematological Malignancies and Solid Tumors

Abstract Number: 2550

A Patnaik[1], M Orloff[2] , R D Carvajal[3], K M Komatsubara[3], C D Britten[4], R Wesolowski[5], G Michelson[6], O Alcantar[6], C Zhang[6], B Powell[6], P Severson[6], ES Martin[6], B Matusow[6] AC Tsiatis[6]
Affiliations: [1]South Texas Accelerated Research Therapeutics, San Antonio, TX, [2]Thomas Jefferson University, Philadelphia, PA, [3]Columbia University Medical Center, New York, NY, [4]Medical University of South Carolina, Charleston, SC, [5]The Ohio State University, Columbus, OH, [6]Plexxikon Inc., Berkeley, CA

Background

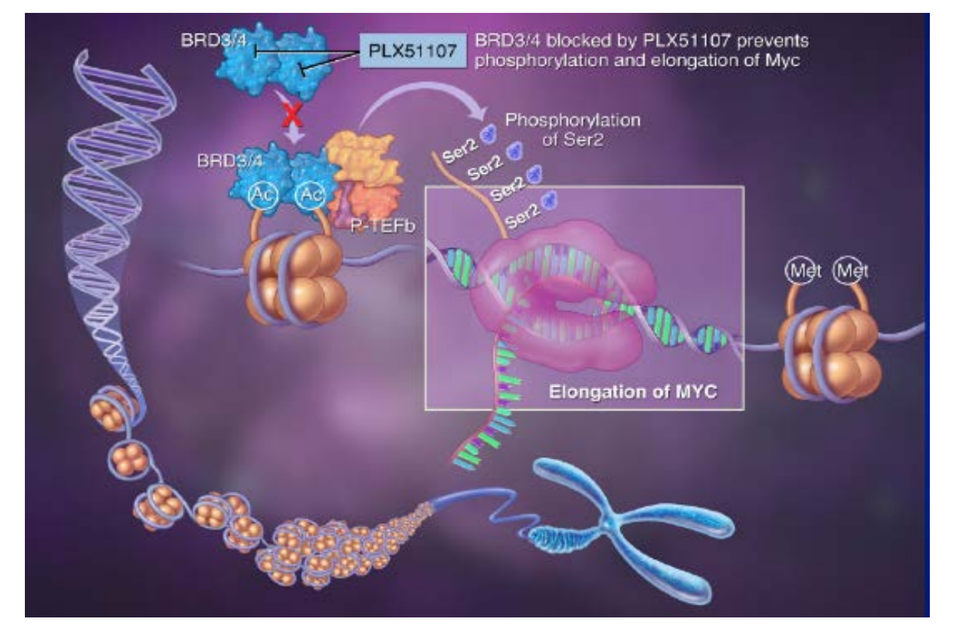
BRD3/4 blocked by PLX51107 prevents phosphorylation and elongation of Myc
PLX51107
Phosphorylation of Ser2
P-TEFb
Elongation of MYC

The BET (bromodomain and extraterminal) subfamily of human bromodomain (BRD) containing proteins consist of four members (BRD2, BRD3, BRD4 and BRDt). Their ability to recognize acetylated histone lysine residues enable these epigenetic readers to modulate gene transcription by recruiting chromatin modifying enzymes and transcription factors to gene promoters and enhancers. The most widely studied example is the oncogene MYC. Additional pharmacodynamics (PD) markers for BET inhibition includes HEXIM1, WDR47, CCR1 and CCR2.

PLX51107 is an orally active small molecule inhibitor with low nanomolar potency in blocking interactions mediated by the four BET family proteins. PLX51107 exhibits anti-tumor activity against a broad panel of solid and liquid tumors cell lines, both *in vitro* and *in vivo*.

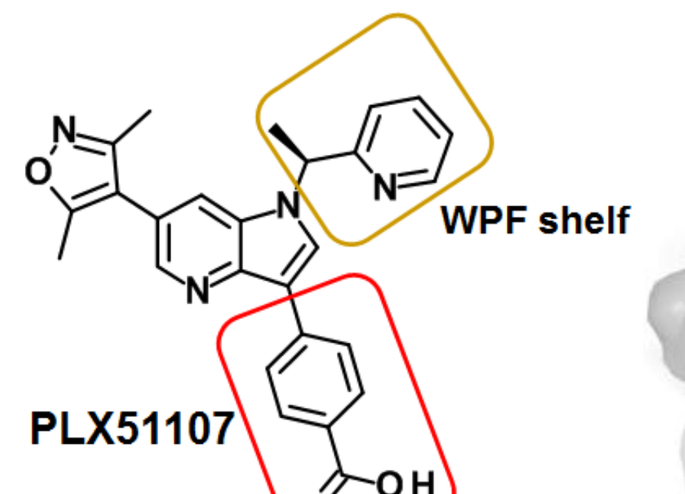
WPF shelf

PLX51107

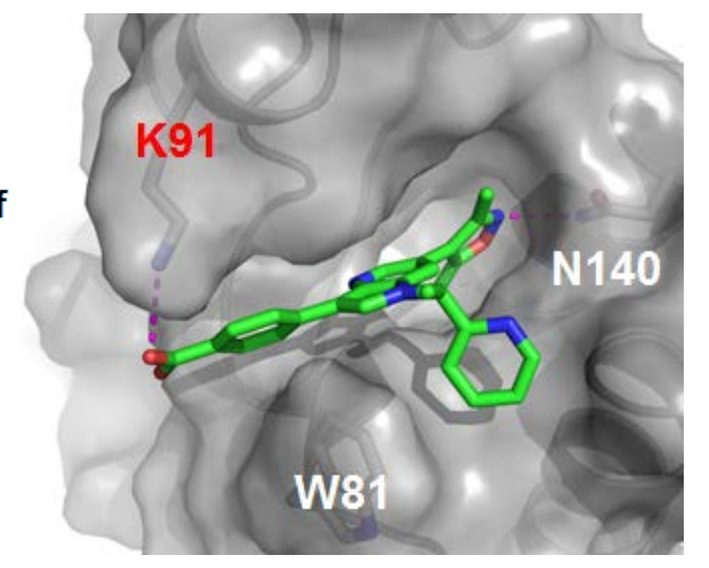
K91
N140
W81

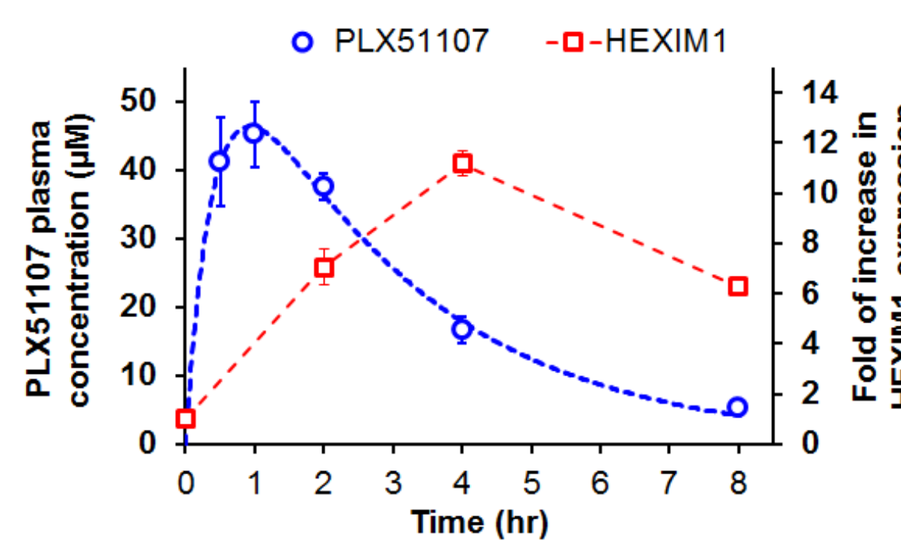
PLX51107
HEXIM1
PLX51107 plasma concentration (µM)
Fold of increase in HEXIM1 expression
Time (hr)

PLX51107 exhibits a unique pharmacokinetic (PK) profile exhibiting rapid absorption and elimination resulting in a high Cmax and short T1/2. A longer lasting PD response can be seen here from xenograft RNAseq analysis (HEXIM1).

Here, we report interim results from the first 8 cohorts of an ongoing Phase 1 study in patients with advanced, relapsed or refractory solid tumors and hematological malignancies.

Trial Design and Methodology

Phase 1b, open-label, multi-dose, dose escalation (NCT02683395):

Objectives:

- Primary: Establish PLX51107 single agent safety and pharmacokinetics (PK)
– Establish single agent maximum tolerated dose/recommended phase 2 dose (MTD/RP2D)
- Secondary: Evaluate efficacy by Objective Response Rate (ORR) by RECIST 1.1 including Duration of Response (DOR) and Progression-Free Survival (PFS)
- Exploratory: Assess changes in gene expression and other biomarkers in peripheral blood & tumor tissue

Methodology:

- Standard 3+ 3 dose escalation for single agent in adults with solid tumor (including lymphoma) or hematological malignancies who have relapsed or refractory disease
- 28 day DLT window –
 - General non-hematologic DLT criteria: any grade ≥ 3 (AE/Lab) toxicity despite adequate supportive care; exceptions made for clinically insignificant or short lived events.
 - Hem DLT: grade 3+ neutropenia, thrombocytopenia; grade 4 anemia.
- PLX51107 was administered orally under fasted conditions QD or BID over 28-day cycle, with continuous dosing

Acknowledgements: The authors would like to thank the patients who participated in this study, as well as the physicians and research staff at START, Thomas Jefferson University, Columbia University, Medical University of South Carolina, and The Ohio State University.

Results

Patient Characteristics and Study Progress

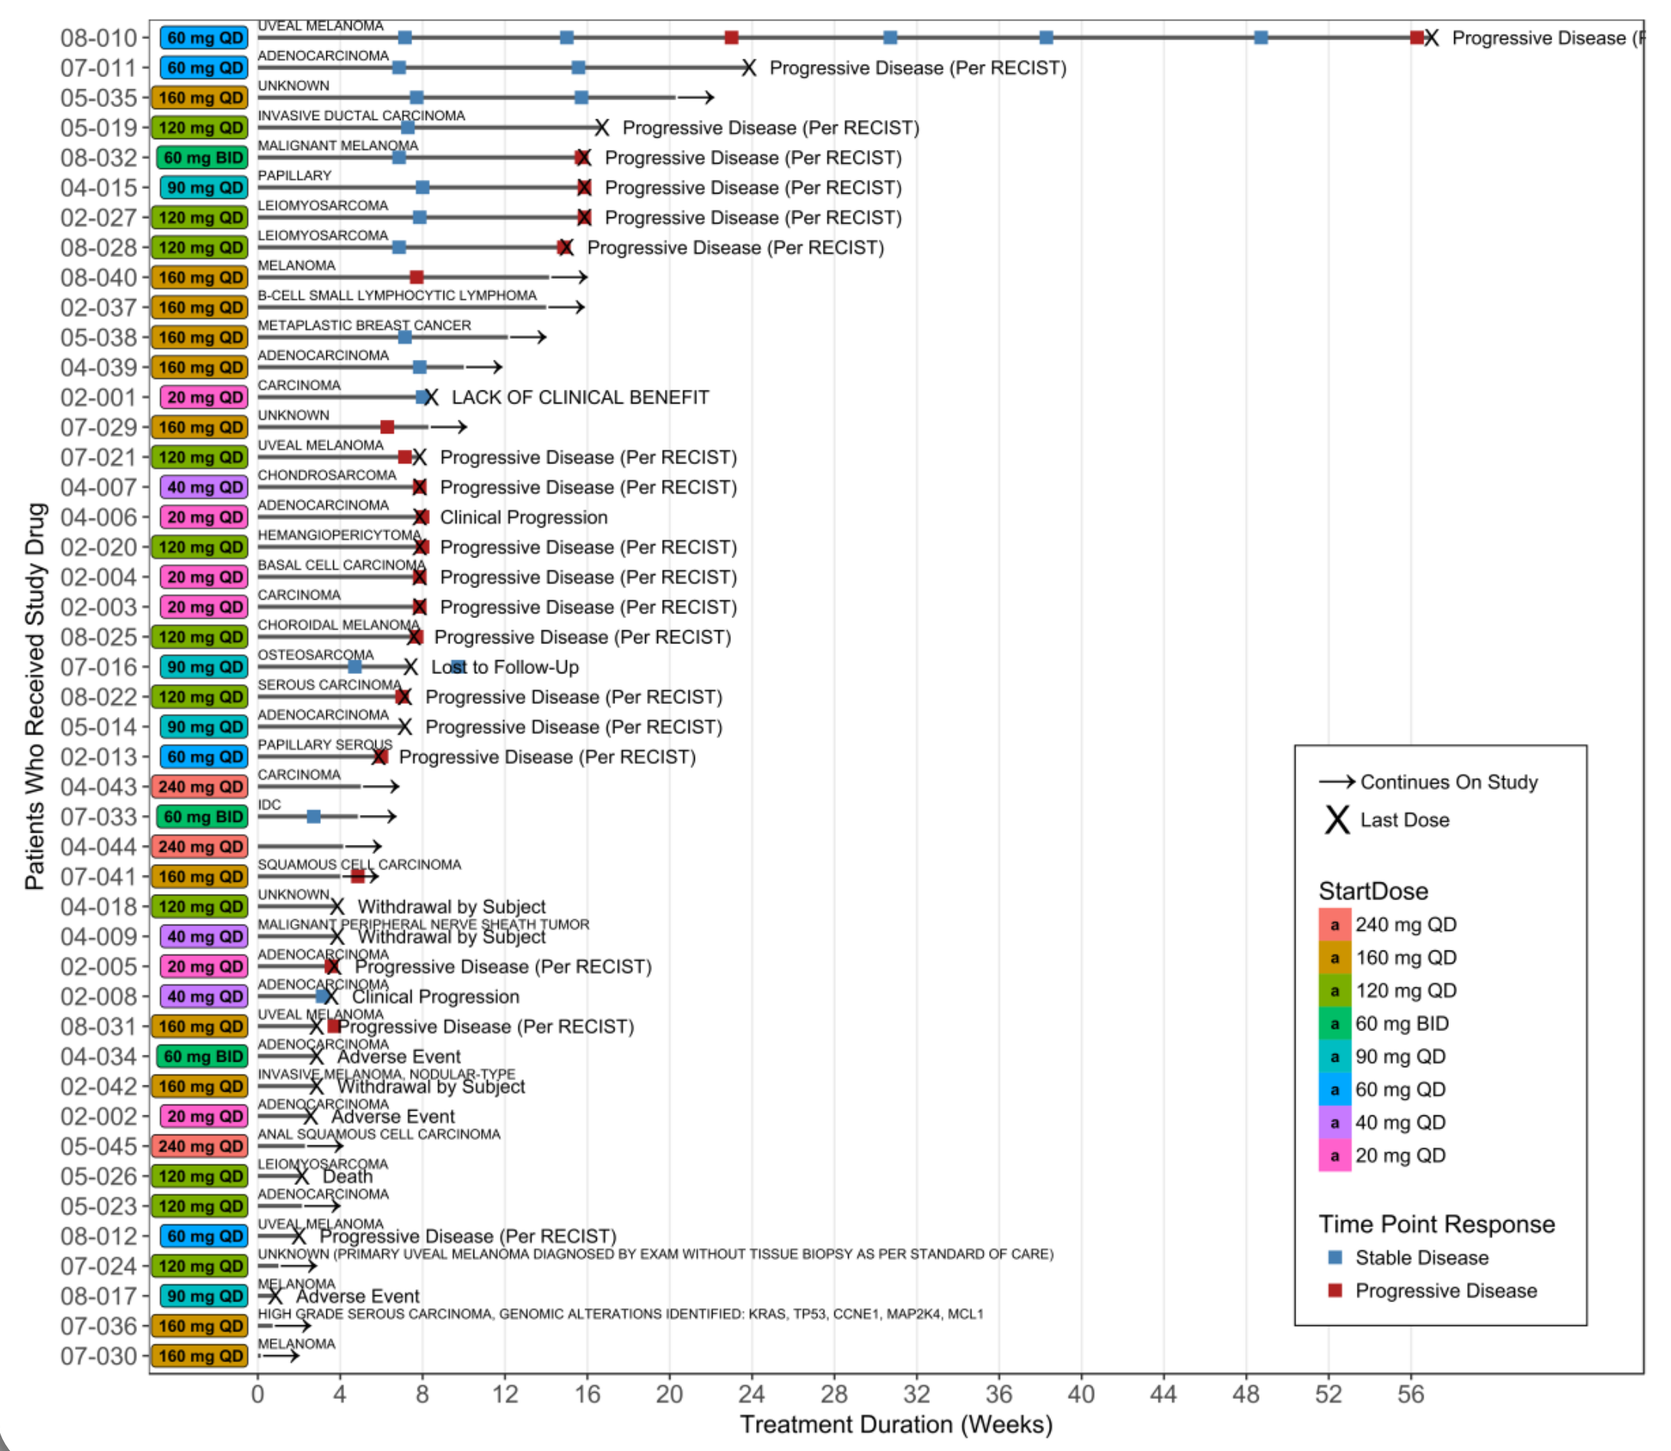
Patients Who Received Study Drug
Treatment Duration (Weeks)
Continues On Study
Last Dose
StartDose
240 mg QD
160 mg QD
120 mg QD
60 mg BID
90 mg QD
60 mg QD
40 mg QD
20 mg QD
Time Point Response
Stable Disease
Progressive Disease

	All Patients (N=45)
Age, median [range]	61 [25-86]
Sex, male, n (%)	17 (37.8)
Prior Therapies, median [range]	7 [3-19]

Starting Dose	Patients Enrolled
20 mg QD	6
40 mg QD	3
60 mg QD	4
90 mg QD	4
120 mg QD	11
60 mg BID	3
160 mg QD	11
240 mg QD	3

Pharmacokinetics

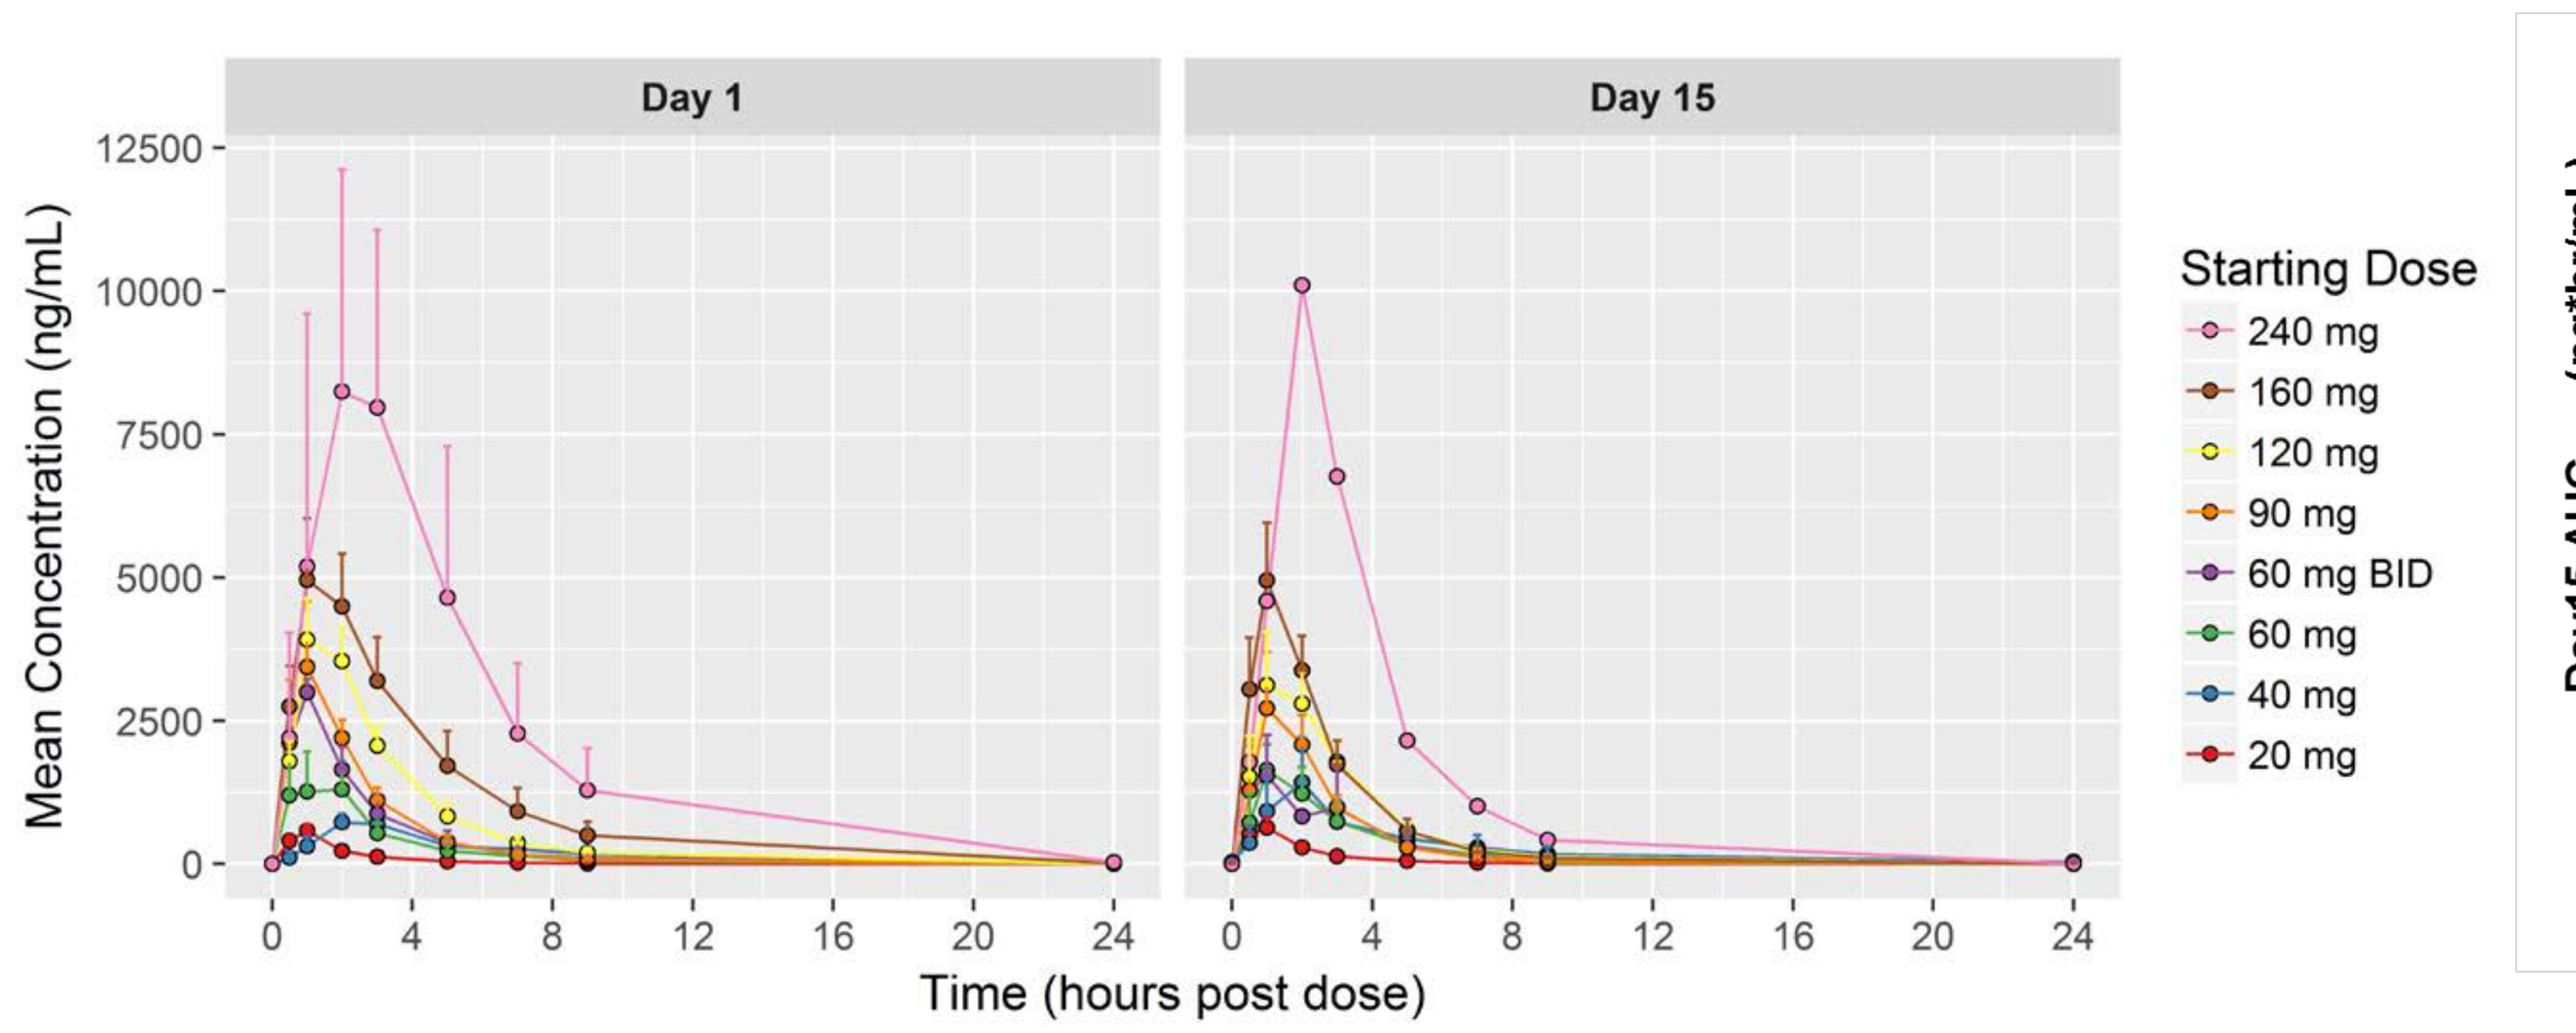
Day 1
Day 15
Mean Concentration (ng/mL)
Time (hours post dose)
Starting Dose
240 mg
160 mg
120 mg
90 mg
60 mg BID
60 mg
40 mg
20 mg

- Rapidly absorbed, high Cmax
- Short half-life (<2.5hr), no accumulation

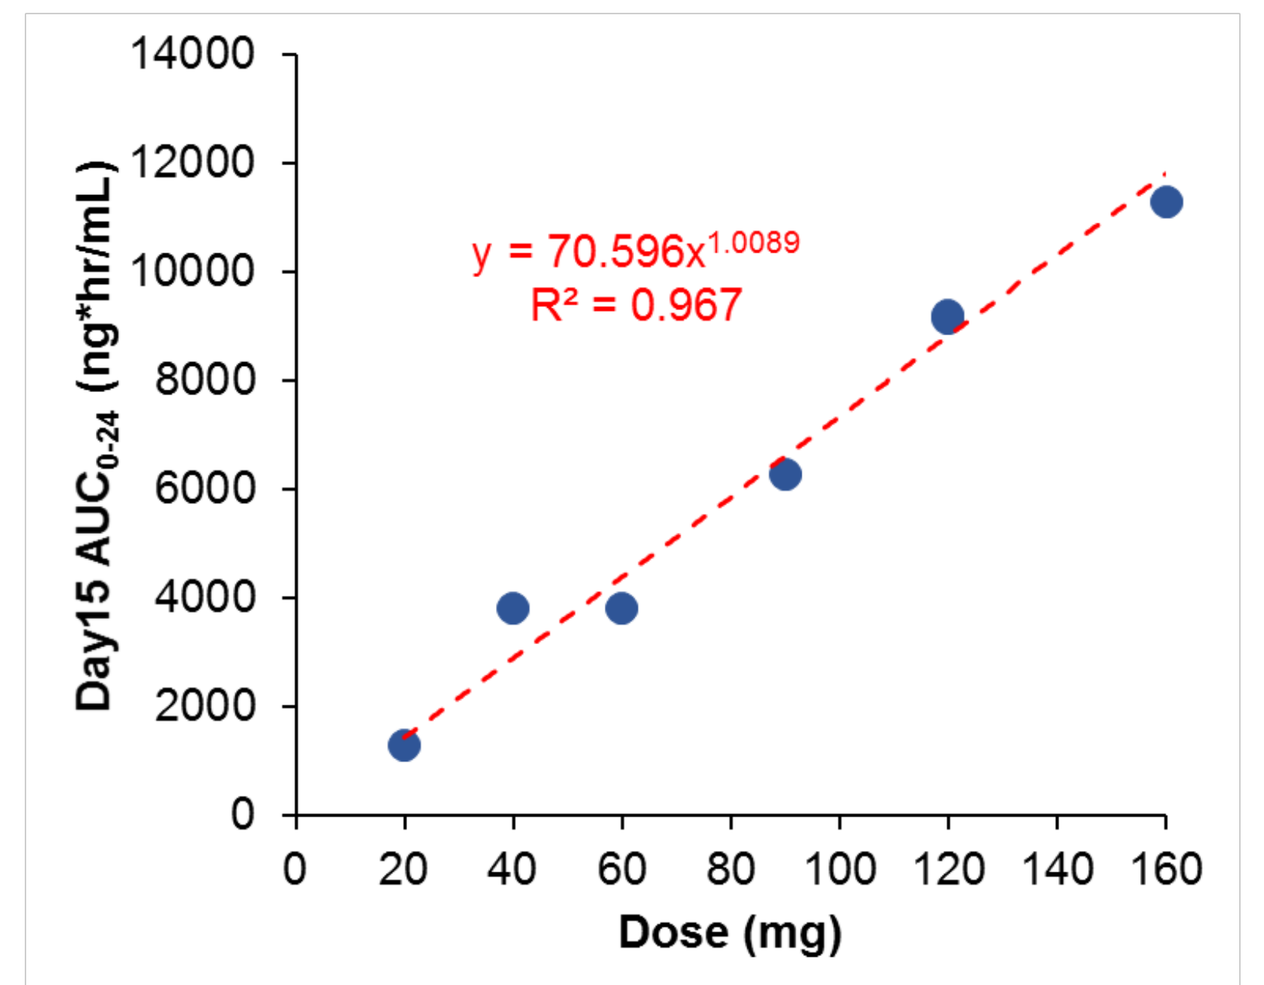
y = 70.596x^1.0089
R² = 0.967
Day15 AUC0-24 (ng*hr/mL)
Dose (mg)

- Dose linear PK from 20 – 160 mg
- 60 mg BID resulted in the same daily AUC as 120 mg QD

Effect on Pro-Survival Pathways in an SLL Patient

Patient 02-037 presented with B-Cell SLL:

- Prior therapies included Cytoxan+Rituximab+Fludarabine, Ibrutinib, Bendamustine and Cemiplimab
- Whole blood gene expression profiling demonstrated that 02-037 had a unique profile including over-expression of key drivers (BCL2, BCL11A, etc.) consistent with a high number of circulating tumor cells.
- On day 1, a single dose of 160 mg PLX51107 temporarily reduced over-expression of pro-survival genes.

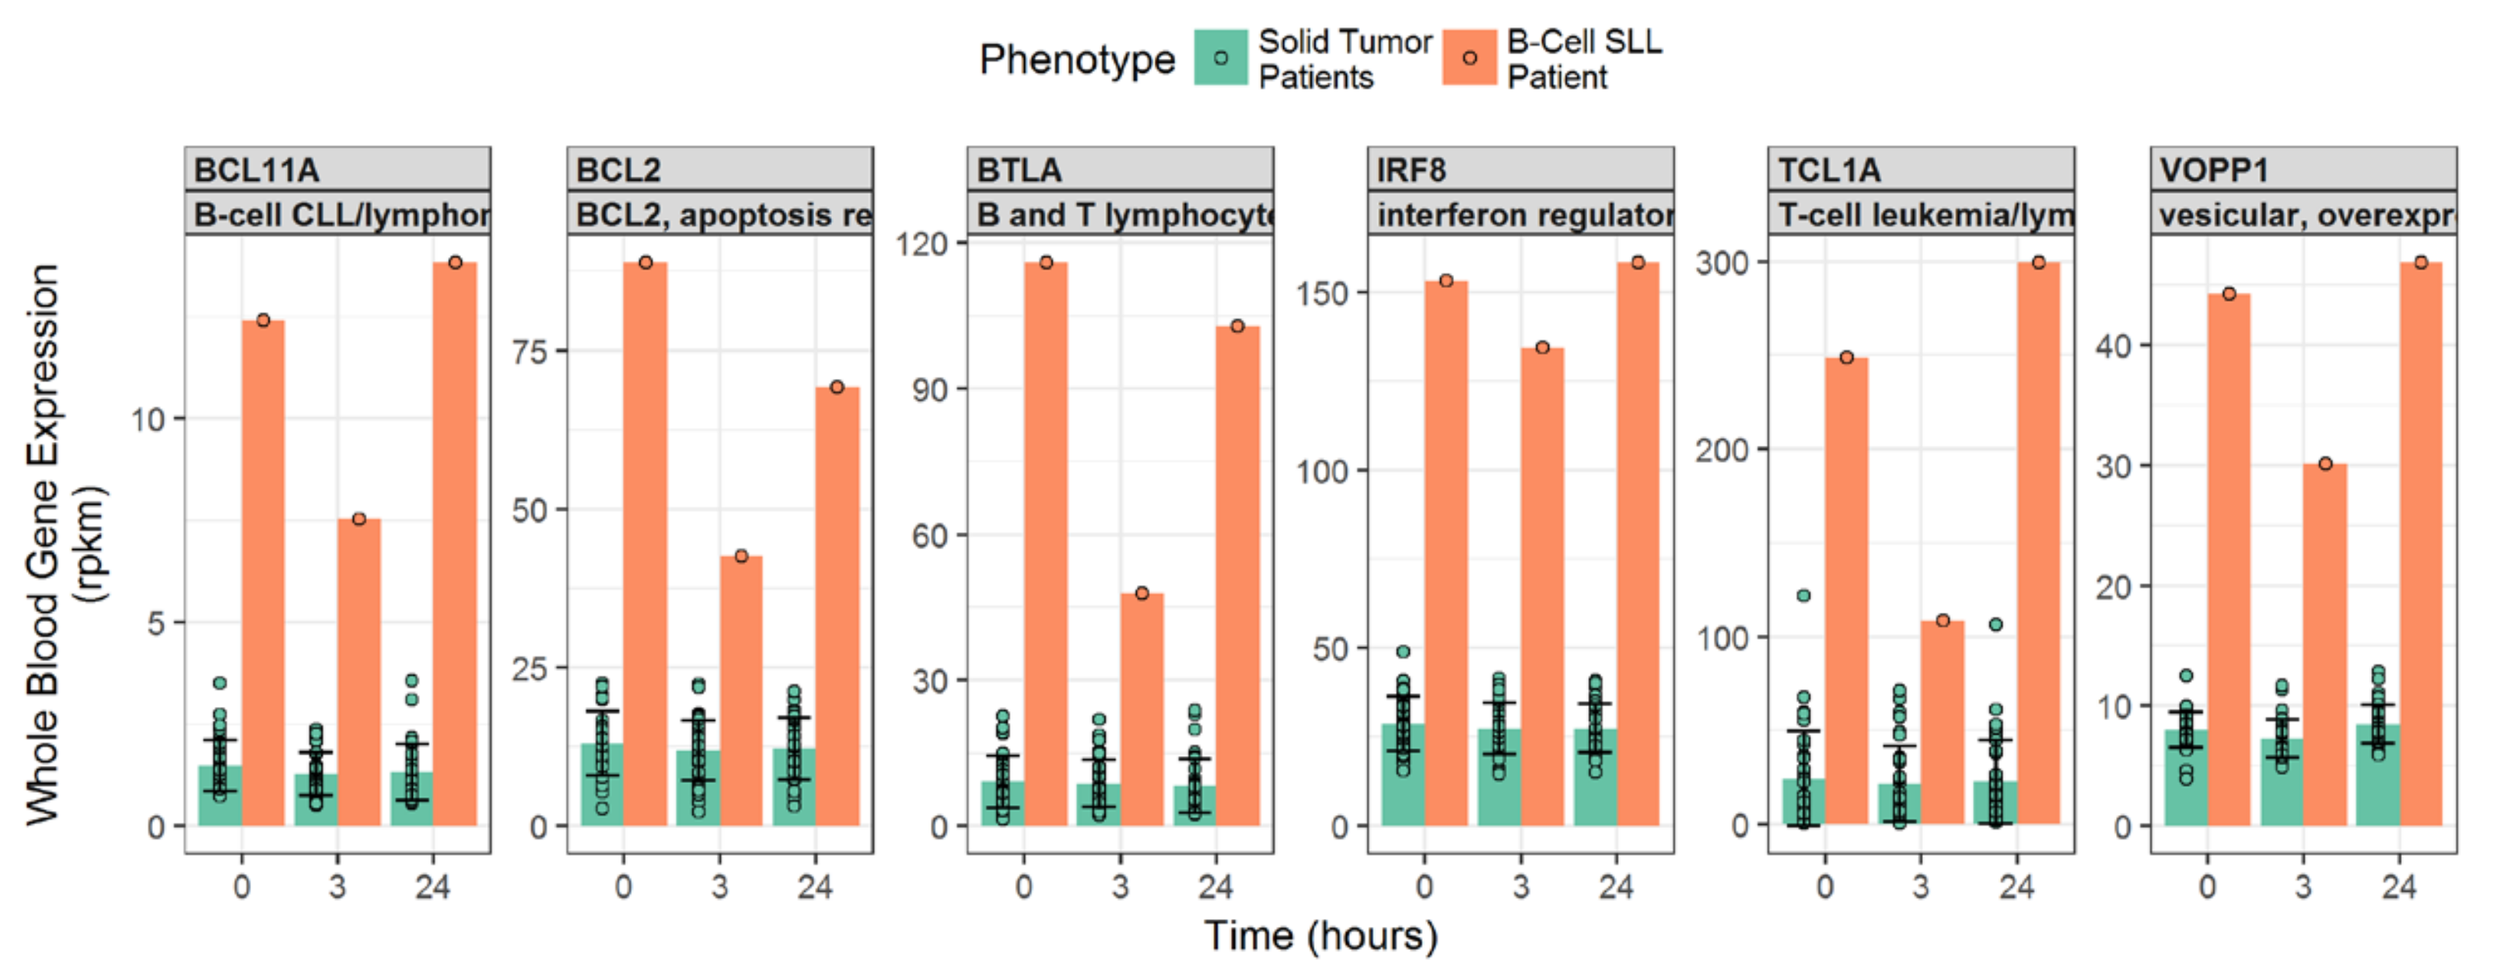
Phenotype
Solid Tumor Patients
B-Cell SLL Patient
BCL11A
BCL2
BTLA
IRF8
TCL1A
VOPP1
Whole Blood Gene Expression (rpkm)
Time (hours)

Adverse Events

AE Term	Grade 1 (Mild)	Grade 2 (Moderate)	Grade 3 (Severe)	All Grades, n (%)
Nausea	14 (31%)	6 (13%)	1 (2%)	21 (47%)
Fatigue	8 (18%)	6 (13%)	0	14 (31%)
Vomiting	9 (20%)	2 (4%)	1 (2%)	12 (27%)
Diarrhea	6 (13%)	4 (9%)	1 (2%)	11 (24%)
Decreased appetite	3 (7%)	4 (9%)	0	7 (16%)
Blood bilirubin increased	4 (9%)	1 (2%)	1 (2%)	6 (13%)
Anemia	1 (2%)	3 (7%)	0	4 (9%)
Dry mouth	3 (7%)	1 (2%)	0	4 (9%)
Myalgia	3 (7%)	1 (2%)	0	4 (9%)
Activated partial thromboplastin time prolonged	4 (9%)	0	0	4 (9%)
Dizziness	4 (9%)	0	0	4 (9%)
Mucosal inflammation	2 (4%)	1 (2%)	0	3 (7%)
Proteinuria	2 (4%)	1 (2%)	0	3 (7%)
Pyrexia	3 (7%)	0	0	3 (7%)
International normalized ratio increased	3 (7%)	0	0	3 (7%)
Muscle spasms	3 (7%)	0	0	3 (7%)
Headache	3 (7%)	0	0	3 (7%)
Thrombocytopenia	0	1 (2%)	1 (2%)	2 (4%)

➢ One Grade 4 AE: Lipase increased

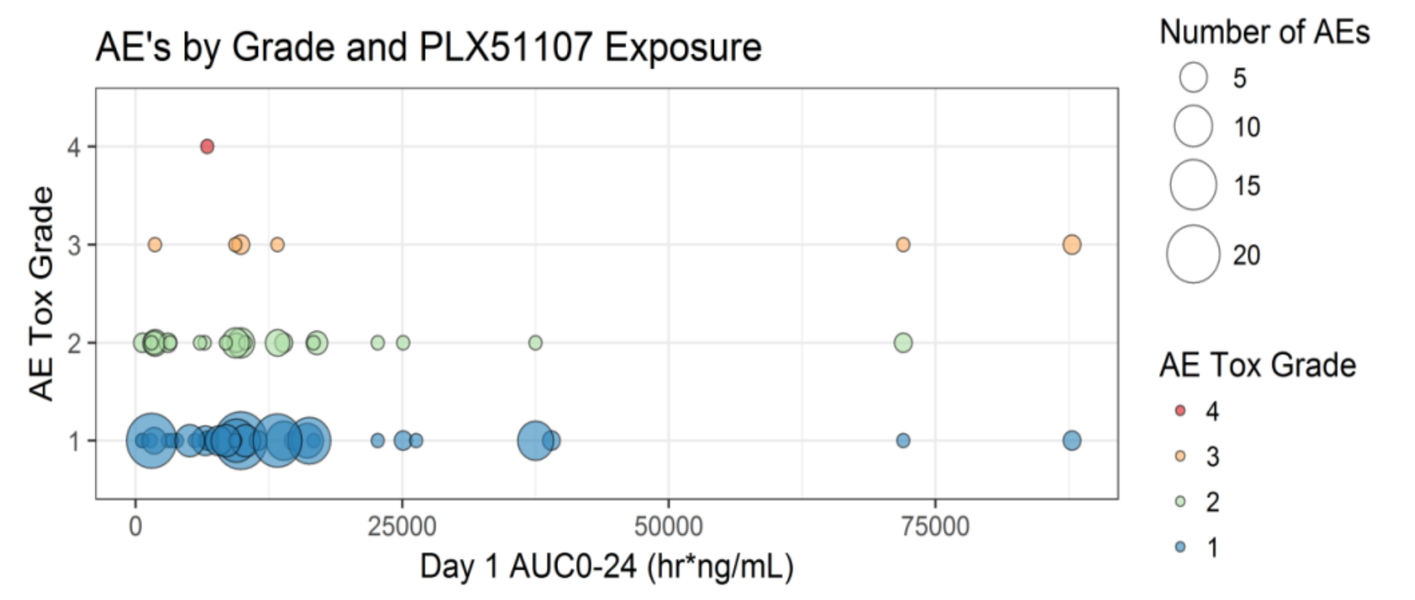
AE's by Grade and PLX51107 Exposure
AE Tox Grade
Day 1 AUC0-24 (hr*ng/mL)
Number of AEs
AE Tox Grade

DLTs:
20 mg QD: (1) G3 Thrombocytopenia
120 mg QD: (1) G3 Nausea/Vomiting
160 mg QD: (1) G2 Acute kidney injury
240 mg QD: (1) G3 Diarrhea,
(1) G2 Hyperbilirubinemia

- **Preliminary MTD appears to be below 240 mg pending completion and analysis of additional study cohort(s)**
- **Overall, the majority of AEs were Grade 1 and consistent with preclinical animal model experience**

Pharmacodynamics (PK/PD)

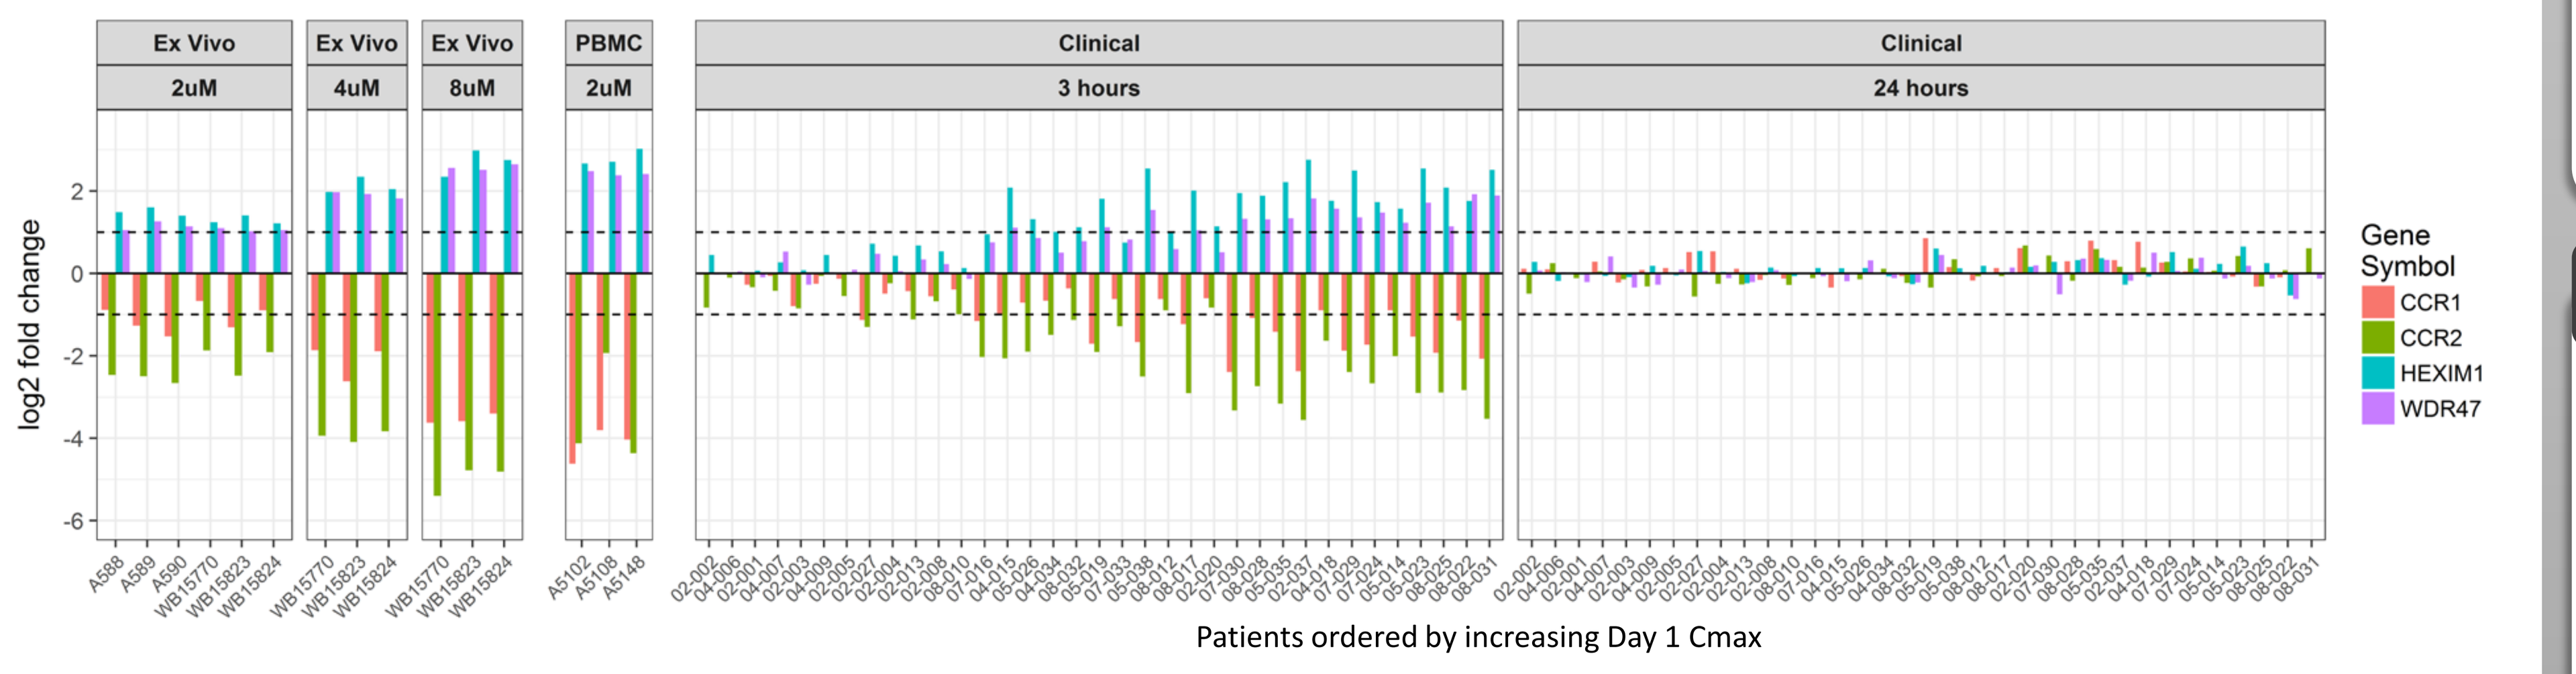
Ex Vivo 2uM
Ex Vivo 4uM
Ex Vivo 8uM
PBMC 2uM
Clinical 3 hours
Clinical 24 hours
log2 fold change
Gene Symbol
CCR1
CCR2
HEXIM1
WDR47
Patients ordered by increasing Day 1 Cmax

Top: Preclinical experiments in whole blood (Ex Vivo) and purified PBMCs demonstrated dose-dependent repression (CCR1, CCR2) and induction (HEXIM1, WDR47) of PD response genes with 2 hours of continuous exposure to PLX51107. In the clinical study, gene expression profiling of blood at baseline, 3 and 24 hours after dosing on day 1 showed evidence of a PD response

Right: PD4 Score (sum of the absolute values of the log2 fold changes over the 4 PD genes) at 3 hours shows a positive and significant correlation with C_{max} and $AUC_{0\text{-}24}$ on Day 1. By 24 hours after dosing, gene expression has returned to near baseline levels.

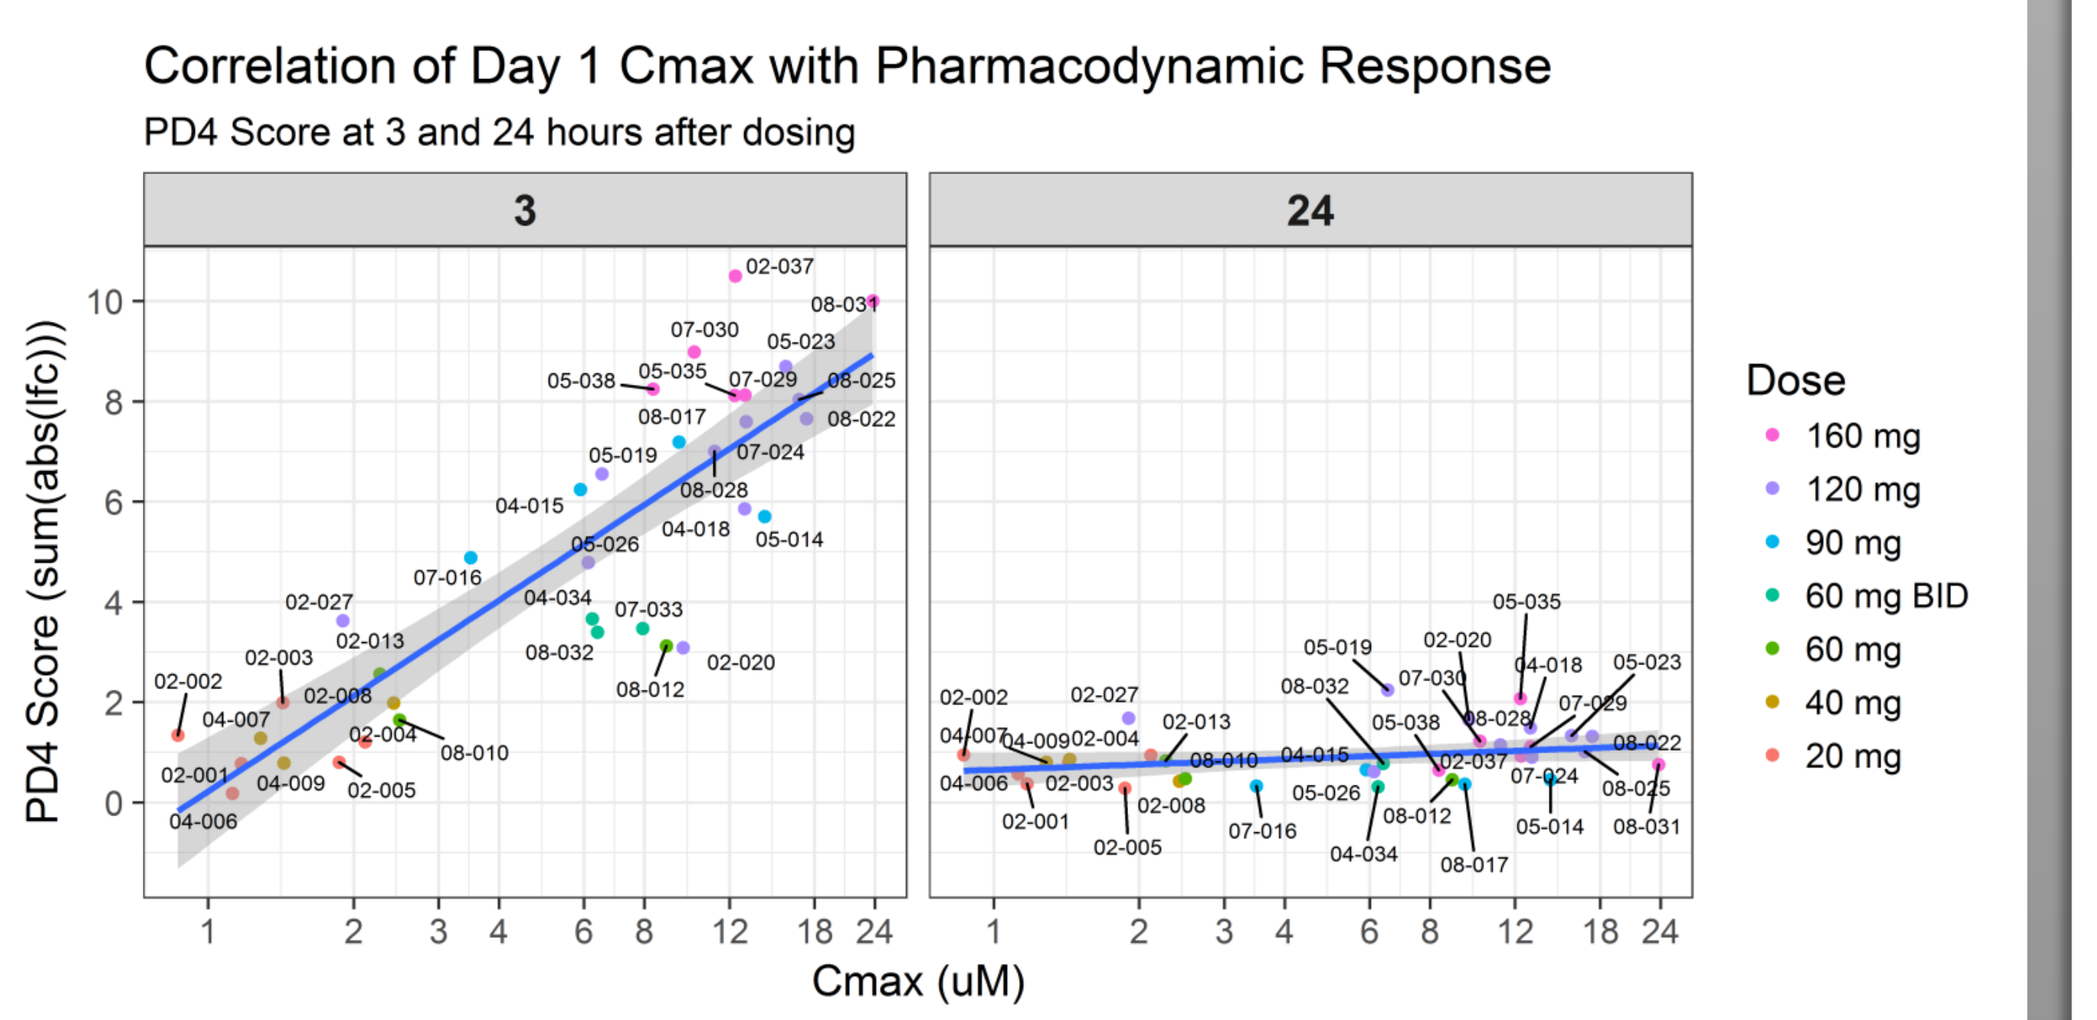
Correlation of Day 1 Cmax with Pharmacodynamic Response
PD4 Score at 3 and 24 hours after dosing
PD4 Score (sum(abs(lfc)))
Cmax (uM)
Dose
160 mg
120 mg
90 mg
60 mg BID
60 mg
40 mg
20 mg

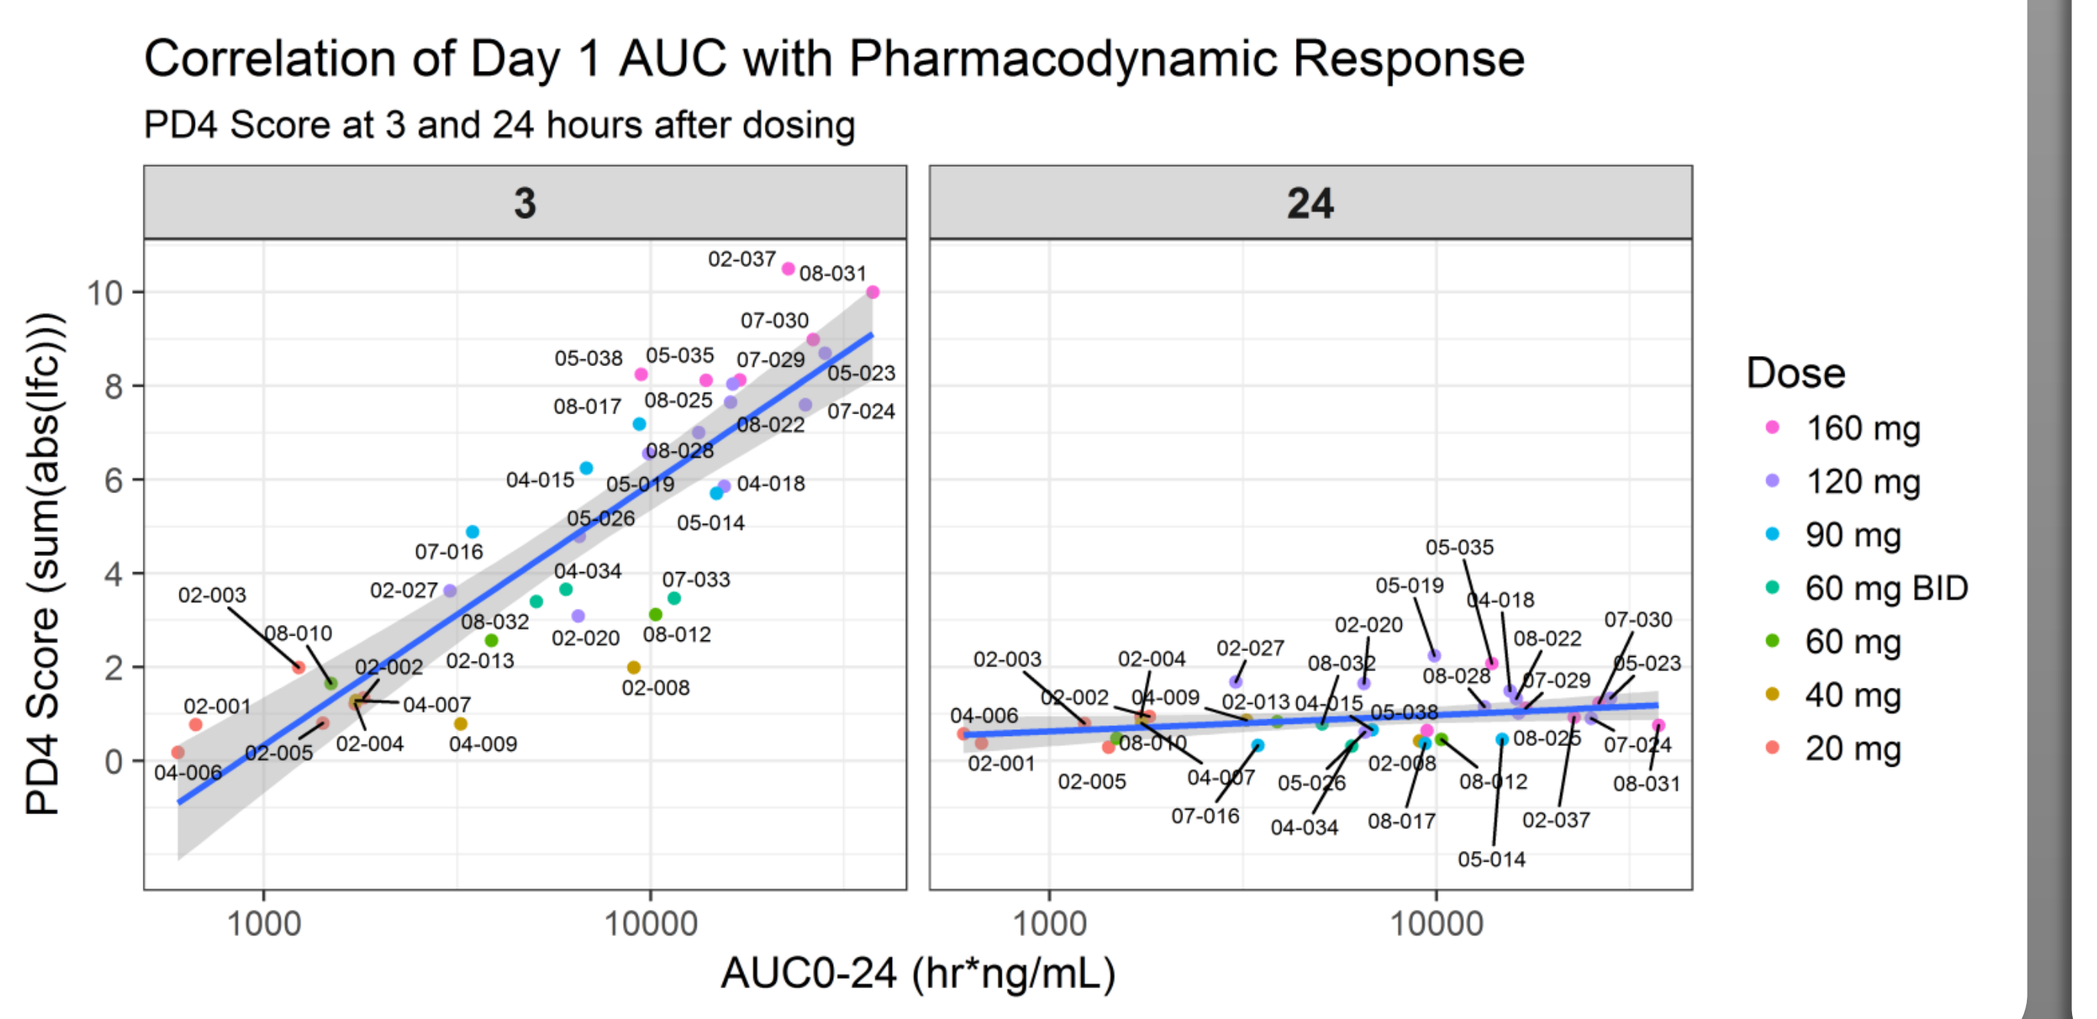
Correlation of Day 1 AUC with Pharmacodynamic Response
PD4 Score at 3 and 24 hours after dosing
PD4 Score (sum(abs(lfc)))
AUC0-24 (hr*ng/mL)
Dose
160 mg
120 mg
90 mg
60 mg BID
60 mg
40 mg
20 mg

Day 1 PK/PD	Correlation of Cmax with PD4 Score		Correlation of AUC0-24 with PD4 Score	
PD Timepoint (Hours)	Pearson R	P-value	Pearson R	P-value
3	0.84	1.90E-10	0.84	1.70E-10
24	0.29	0.095	0.27	0.12

Conclusions

- **PLX51107 is a structurally distinct BET inhibitor which is well tolerated and shows evidence of BET inhibition in patients**
- **Varying dose levels were tested ranging from 20 mg to 240 mg QD**
- **PK was dose linear from 20 mg to 160 mg; (with the same daily dose) BID dosing resulted in the same exposure as QD dosing**
- **Based on interim PK and animal model data, the anticipated RP2D will be between 160 and 240 mg**
- **PLX51107 is rapidly absorbed, resulting in a short half-life (<2.5hr) and exhibits a unique PK profile achieving high Cmax**
- **RNA profiling provided evidence of a marked, dose-dependent, pharmacodynamic response indicative of BET inhibition**
- **PLX51107 was shown to temporarily reduce malignancy associated over-expression (BCL2, BCL11A, etc.) in lymphoid-associated circulating tumor**
- **Further development of PLX51107 as single agent and in combination with other agents in solid tumors, AML/MDS and other hematological malignancies may be considered**
- **Data from this study will be used to inform the ongoing study of PLX2853, a newer and more potent agent in advanced solid tumors and AML/MDS (NCT03297424)**